A Pilot Study Utilizing Molecular Profiling to Find Potential Targets and Select Individualized Treatments for Patients with Metastatic Breast Cancer

Gayle Jameson[1], Emanual Petricoin[3], Jasgit Sachdev[1], Lance Liotta[3], David Loesch[5], Stephen Anthony[4], Manpreet Chadha[1], Mariaelena Pierobon[3], Kimberly Reeder[3], Monica Fulk[1], Linda Vocila[6], Nina Cantafio[6], Bryant Dunetz[7], Nicholas Robert[2]

[1]Virginia G Piper Cancer Center Clinical Trials, Scottsdale Healthcare, Scottsdale, AZ . [2]Virginia Cancer Specialists, US Oncology, Fairfax, VA . [3]Center for Applied Proteomics and Molecular Medicine (CAPMM) at George Mason University, Manassas, VA . [4]Evergreen Hematology & Oncology, Spokane, WA . [5]Caris Life Sciences, Phoenix, AZ . [6]Translation Drug Development, (TD2), Scottsdale, AZ . [7]The Side-Out Foundation, Fairfax, VA

BACKGROUND

Historical Perspective

A recent non-randomized pilot study utilizing molecular profiling (MP) of patients' tumors to find potential targets and select treatments (Von Hoff, et al., 2010), concluded that:

- This MP approach to selecting treatments benefitted 27% of patients (n=66) with a variety of solid tumor types (95% CI 17-38%) p=0.007, who had disease progression on all prior therapies.
- Clinical benefit was defined by a **Growth Modulation Index (GMI) of ≥ 1.3 which calculated the patient's PFS ratio (PFS on MP selected therapy/PFS on prior therapy).**
- **44% (8 of 18) patients with metastatic breast cancer demonstrated longer PFS on an MP suggested regimen compared with the most recent prior regimen on which the patient had experienced PD, (GMI of ≥ 1.3).**
- Initiating disease specific trials and introducing new MP methodologies are warranted.
- Current study was based on the methods and design of Von Hoff, et al., 2010 and added novel RPMA tumor analysis.

OBJECTIVES

Primary

To determine the percent of patients with refractory breast cancer where MP and RPMA based protein pathway activation analysis of their tumor, can change the clinical course of their disease (i.e. produce a Growth Modulation Index (GMI) ≥1.3).

Secondary

To determine:

- Frequency with which MP analysis of a patient's tumor by IHC/FISH and/or microarray and RPMA yields a target against which there is a commercially available, approved agent or therapeutic regimen.
- Percent of time in which the treatment selected by MP and RPMA analysis is different than that which would have been selected by the patient's physician.
- Response rate (according to RECIST and tumor marker-specific response criteria).
- Overall survival, in patients whose therapy is selected by MP and RPMA.
- Optimal tissue biomarker subset that is the most predictive of a given therapy leading to an improved outcome (as measured by GMI and response rate).

STUDY DESIGN

- Open-label, multicenter pilot study

Patient Eligibility

Key inclusion criteria:

- ≥18 years of age
- ECOG: 0-1
- Patients with a diagnosis of metastatic breast cancer, measurable or evaluable non-measurable disease (lesions below the limits defined for measurable disease in RECIST 1.1)
- Refractory disease as defined below:
 - Progression of disease (PD) on ≥ 3 prior chemotherapeutic or biological regimens for advanced disease
 - PD on the last treatment or within 2 months of the last treatment dosing
 - Received ≥ 4 weeks but ≤ 6 months of the last treatment

STUDY DESIGN (cont.)

Study Schema

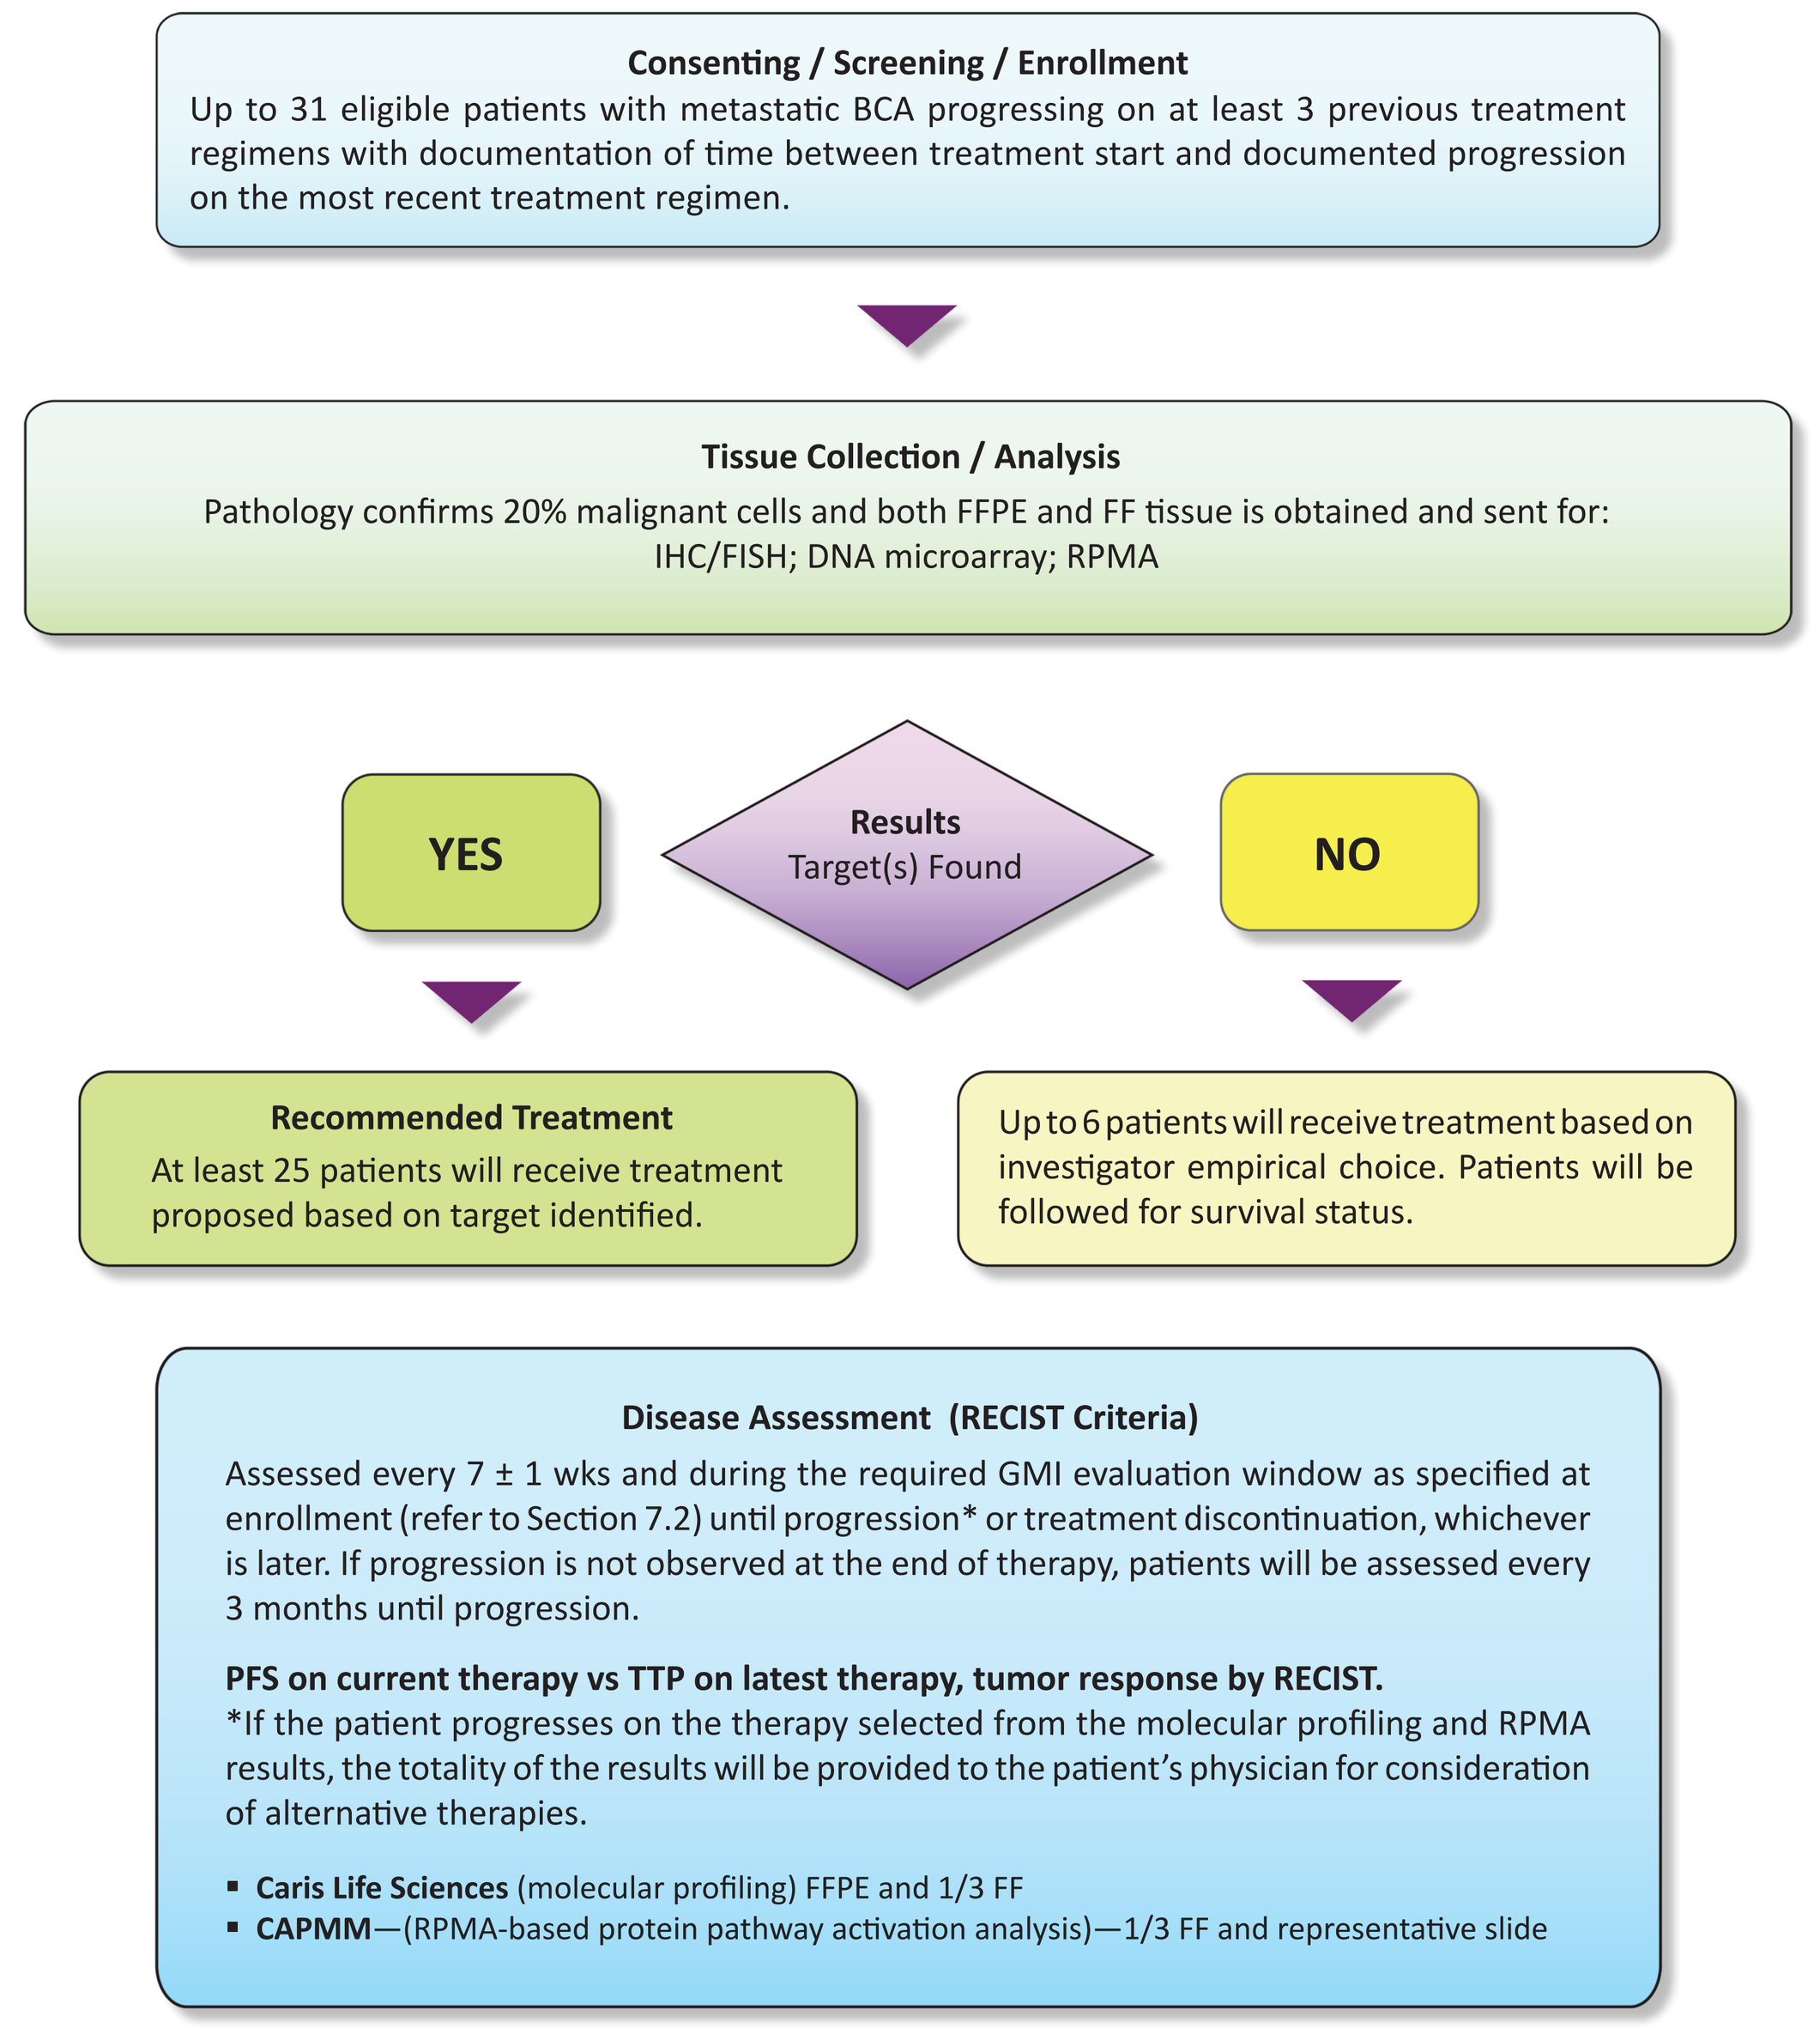
Consenting / Screening / Enrollment
Up to 31 eligible patients with metastatic BCA progressing on at least 3 previous treatment regimens with documentation of time between treatment start and documented progression on the most recent treatment regimen.
Tissue Collection / Analysis
Pathology confirms 20% malignant cells and both FFPE and FF tissue is obtained and sent for: IHC/FISH; DNA microarray; RPMA
YES
Results Target(s) Found
NO
Recommended Treatment
At least 25 patients will receive treatment proposed based on target identified.
Up to 6 patients will receive treatment based on investigator empirical choice. Patients will be followed for survival status.
Disease Assessment (RECIST Criteria)
Assessed every 7 ± 1 wks and during the required GMI evaluation window as specified at enrollment (refer to Section 7.2) until progression* or treatment discontinuation, whichever is later. If progression is not observed at the end of therapy, patients will be assessed every 3 months until progression.
PFS on current therapy vs TTP on latest therapy, tumor response by RECIST.
*If the patient progresses on the therapy selected from the molecular profiling and RPMA results, the totality of the results will be provided to the patient's physician for consideration of alternative therapies.
Caris Life Sciences (molecular profiling) FFPE and 1/3 FF
CAPMM—(RPMA-based protein pathway activation analysis)—1/3 FF and representative slide

Tumor Analysis

- IHC/FISH, DNA microarray (Caris Life Sciences, Phoenix, AZ)
- Reverse Phase Protein Microarray (RPMA); Measures levels of activation of phosphorylation of target proteins (CAPMM at George Mason University)

Reverse Phase Protein Microarray Work-flow

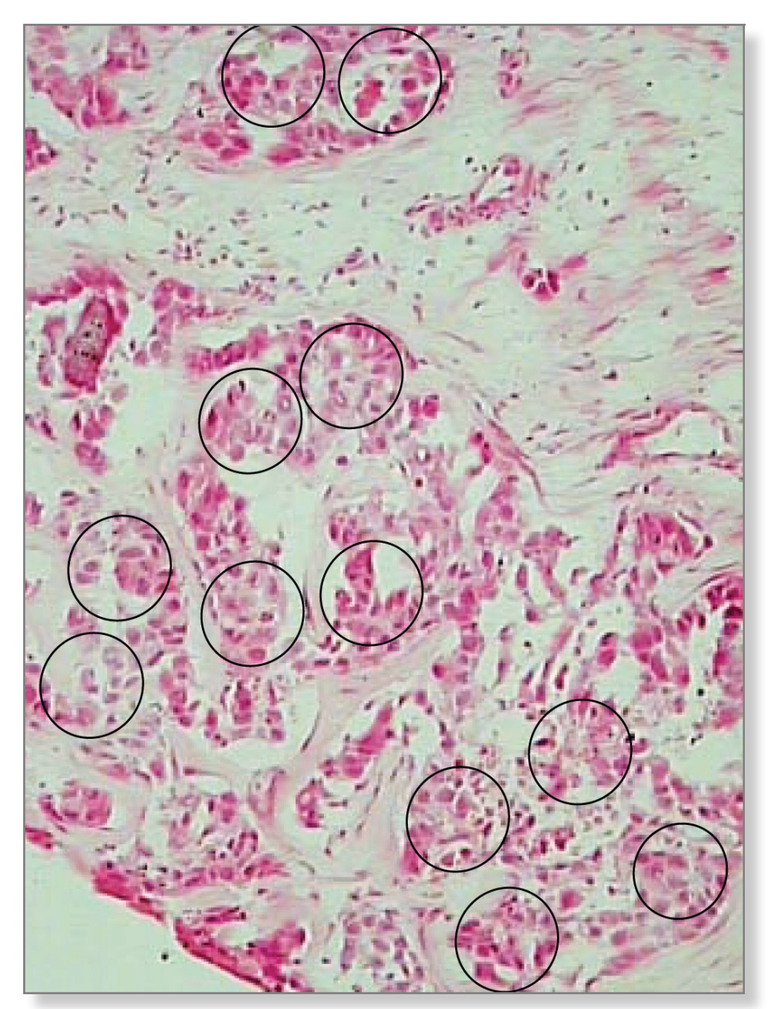

Tumor cells were isolated using Laser Capture Microdissection technology. Samples and controls were printed in triplicate onto nitrocellulose slides.

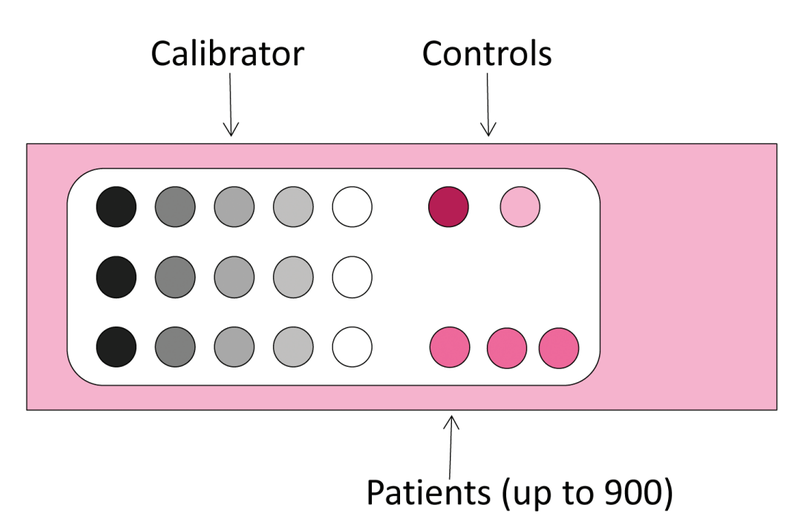
Calibrator
Controls
Patients (up to 900)

Arrays were tested with 11 antibodies against FDA-approved drug targets and downstream substrates.

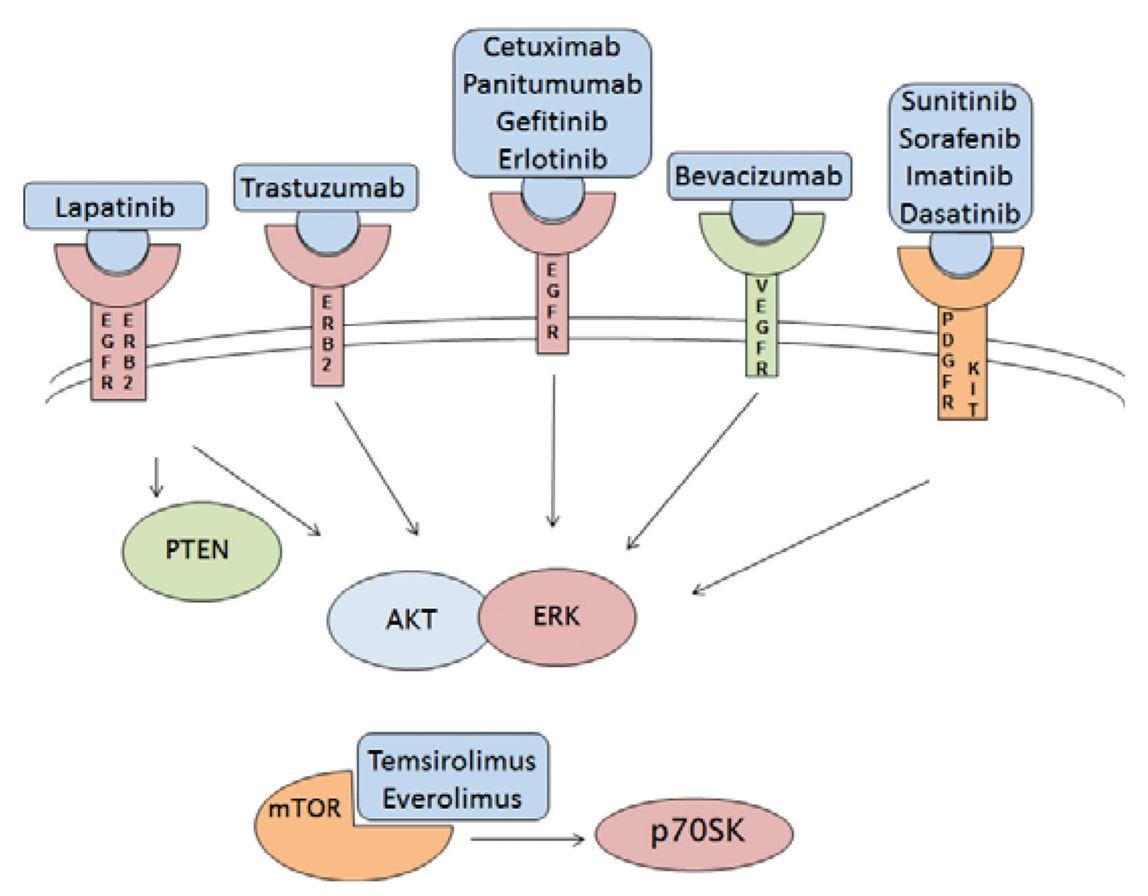
Lapatinib
Trastuzumab
Cetuximab
Panitumumab
Gefitinib
Erlotinib
Bevacizumab
Sunitinib
Sorafenib
Imatinib
Dasatinib
EGFR
ERB2
EGFR
VEGFR
PDGFR
KIT
PTEN
AKT
ERK
Temsirolimus
Everolimus
mTOR
p70SK

Treatment Selection Process

A Study Treatment Selection Committee composed of a nurse practicioner, physicians and bench scientists reviewed the results of the three methodologies used and suggested a specific therapy for the patient on the basis of the identified tumor targets and the patient's history.

Algorithm for Treatment Selection

Order of Precedence	Methodology
1	IHC /FISH, and DNA Microarray, or RPMA and DNA Microarray
2	IHC /FISH, or RPMA
3	IHC /FISH, DNA Microarray
4	RPMA alone
5	IHC / /FISH alone
6	RPMA and DNA Microarray
7	DNA Microarray alone

The above algorithm was used in addition to the patient's past medical and treatment history, current AE's from prior therapies e.g. peripheral neuropathy, etc., in selecting the MP recommended treatment.

Patient Characteristics

Characteristic	n	%
Gender		
Female	25	100
Age, Years		
Median	58	
Range	42-77	
Ethnicity		
Not of Hispanic/Latin Origin	24	96
Hispanic/Latin Origin	1	4
Race		
White	24	96
Unknown	1	4
ECOG		
0	15	60
1	10	40
Metastatic at Diagnosis	7	28
Number of Prior Treatment Regimens (Range 4-12)		
4	3	12
5	5	20
6	3	12
7	4	16
8	5	20
9	3	12
11	1	4
12	1	4
Tumor Characteristics at Diagnosis		
ER		
Positive	22	88
Negative	3	12
PR		
Positive	13	52
Negative	12	48
Her2		
Positive	6	24
Negative	17	68
Unknown	2	8
BRCA-2	2	8

Patient Summary

- **Enrolled = 28**
- **Completed tumor biopsy = 28**
- **Treated on study / Evaluable = 25**
 3 patients not treated on study.
 2 palliative care; 1 received treatment off study.
- **Treatment based on MP results = 25**
- **Number of days on MP selected therapy = 9 - 891+**

Adverse Events

- **No unexpected treatment related adverse events - consistent with known AE's of FDA approved agents**
- **No treatment related deaths**
- **Serious adverse events related to protocol required tumor biopsy = 2 Liver hematoma (1), RUQ pain (1)**

RESULTS

Individual Hormone Receptor and HER2 Status, GMI Response, Selected Targets and Treatments

Subject ID	Baseline HR/ HER2 Status	Actual Patient GMI	TTP on Study Treatment (days)	TTP on Last Line of Therapy (days)	Targets Used to Select Treatment and Method Used	Selected Treatment Based on Pt's Tumor MP	Change in HR/ HER2 from Original DX
100	HR + HER2 +	0.459	56	122	ER^{I}, TS^{I}, TYMS^{M} Her2^{I}	Lapatinib+ Capecitabine	
101	HR + HER2 -	1.977	87	44	ER^{I}, PR^{I}, TOPO1^{I} TOP1^{M}	Irinotecan	
102	HR - HER2 -	0.465	20	43	HER2^{I}, ERBB2^{M}, PTEN^{I}	Lapatinib+ Herceptin	HER2 +
103	HR - HER2 -	0.629	44	70	ER^{I}, PR^{I}, AR^{I}, PTEN^{I}, EGFR^{F}	Erlotinib+ Letrozole	ER +, PR +
104	HR + HER2 -	0.236	38	161	TS^{I}	Capecitabine	
105	HR + HER2 -	0.319	15	47	TOPO^{I}, TOP1^{M}	Irinotecan	
107	HR + HER2 -	1.303	86	66	TOPO1^{I}, TS^{I}	FOLFIRI	
108	HR + HER2 -	1.181	196	166	TOP2A^{I,M}, PGP^{I}	Doxil	
109	HR + HER2 -	5.156	892	173	TOP2A^{I,M}, Her2^{I,F}, PTEN^{I}	Irinotecan + Trastuzumab	HER2 +
110	HR + HER2 +	6.873	378	55	TOP2A^{I,M}, PGP^{I}, HER2^{I,F}, PTEN^{I}, TS^{I}, TYMS^{M}, TOPO1^{I}	Doxil -> Doxorubicin -> FOLFIRI^{b}	
111	HR + HER2 -	0.857	36	42	TOP2A^{I,M}, PGP^{I}	Doxil	
112	HR + HER2 -	0.046	9	196	TOPO1^{I}, TS^{I}	FOLFIRI	
113	HR + HER2 -	2.260	113	50	ER^{I}, ESR1^{M}, PR^{I}, TOPO1^{I}	Aromasin + Irinotecan	
114	HR + HER2 -	0.729	62	85	ER^{I,} ESR1^{M,} PR^{I}	Letrozole	
115	HR + HER2 -	1.684	165	98	ER^{I}, ESR1^{M}, PR^{IM}, TYMS^{M}, TOPO1^{I}	Letrozole -> FOLFIRI^{C}	
116	HR - HER2 -	2.778	275	99	SPARC^{I}	Abraxane	HR +
117	HR + HER2 -	3.408	351	103	TOPO1^{I}, TOP1^{M}	Irinotecan	
118	HR + HER2 -	2.527	235	93	TOPO1^{I}, TS^{I}	FOLFIRI	
119	HR + HER2 +	1.977	85	43	RRM1^{I,M}, RRM2B^{M}, JRT2^{I,F}, PTEN^{I}, PIK3CA^{I}	Gemcitabine + Trastuzumab	
120	HR + HER2 -	0.170	29	171	AR^{I}	Flutamide	
121	HR + HER2 unk	0.235	20	85	PTEN^{I}, EGFR^{I}, PDGFRA^{M}	Erlotinib	
124	HR + HER2 +	2.783	167	60	EGFR/AKT/ERK pathway activated PTEN^{I}, ER^{I}, ESR1^{M}	Tarceva + Letrozole	
125	HR + HER2 +	2.622	97	37	HER2^{I,F}, PTEN^{I}, TELE3^{I}, SPARC^{I}	Lapatinib + Paclitaxel	
126	HR + HER2 -	0.448	56	125	TOPO1^{I}	Irinotecan	
128	HR + HER2 unk	1.656	106	64	TOPO1^{I}, TS^{I}	XELIRI	

By IHC analysis, ALL patients' tumor samples demonstrated low or absent TS, normal PTEN, increased TOPO1 and MRP1.

Highlight = Positive Outcome with GMI ≥ 1.3

Abbreviations: MP= Molecular Profiling; I=Immunohistochemistry; M=Microarray; R=RPMA; FOLFIRI=Irinotecan + 5FU + Folinic Acid; XELIRI=Xeloda + Irinotecan; unk = unknown

[a] Multiple druggable targets noted on all patients. Only those supporting the treatment selected are listed.

[b] Due to intolerance of Doxil, then reaching lifetime max of anthracylcine, regimen changed FOLFIRI based on MP results
[c] Due to intolerance of Letrozole after 1 week, treatment changed to FOLFIRI

RPMA Results

Drug Target(s)	EGFR Y1173	Erb2 Y1248	VEGFR Y996	PDGFR Y751 cKit Y719 cAbl T735	mTOR S2481
Downstream	ERK	ERK	ERK	ERK	p70SKT389
Substrate from Drug Target	T202/Y204 AKT S473	T202/Y204 AKT S473	T202/Y204 AKT S473	T202/Y204 AKT S473	
Number of	13/25	3/25	0	3/25	0
Pathway Activated Positive Patients	(52.0%)	(12.0%)		(12.0%)	

Molecular profile was not available for 3 of the 25 patients due to inadequate material

Physician first choice of patient's next treatment compared with MP selected therapy: 0 of 25 matches.

Comparison of PFS on MP therapy vs. PFS on prior therapy for 13 patients with GMI of ≥ 1.3

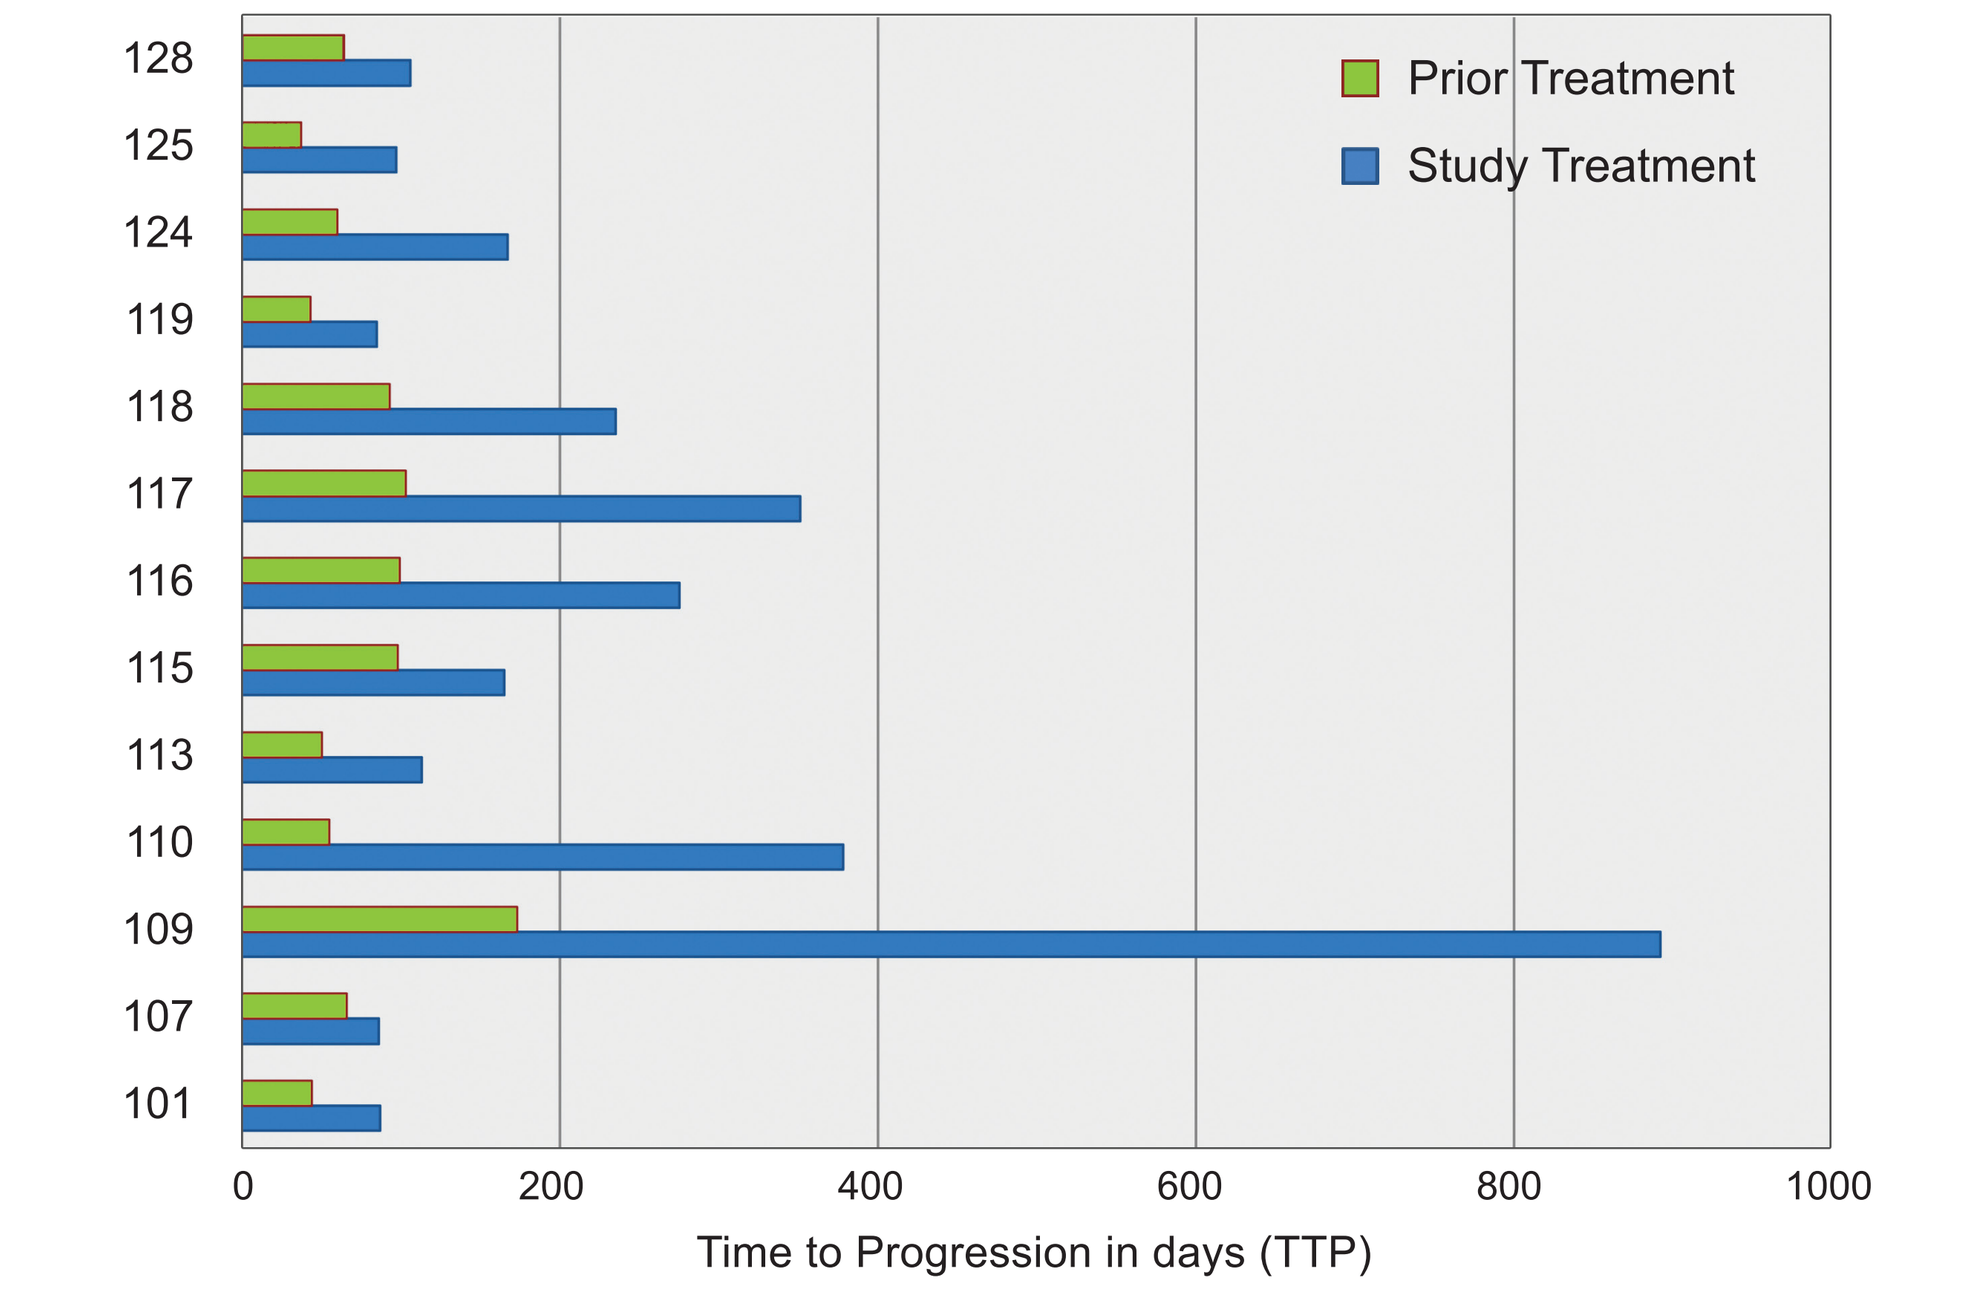
128
125
124
119
118
117
116
115
113
110
109
107
101
Prior Treatment
Study Treatment
0
200
400
600
800
1000
Time to Progression in days (TTP)

Response Rate by RECIST 1.1

Evaluable Patients N = 21

Complete Response (CR) = 0

Partial Response (PR) = 6

Stable Disease (SD) = 12

Progression of Disease (PD) = 3

No restaging scans done due to clinical progression = 4

Disease control rate PR+CR+SD = 18 of 25 total patients = 72%

Maximum percent change of summed diameters of target lesions with respect to baseline diameters by RECIST 1.1 criteria

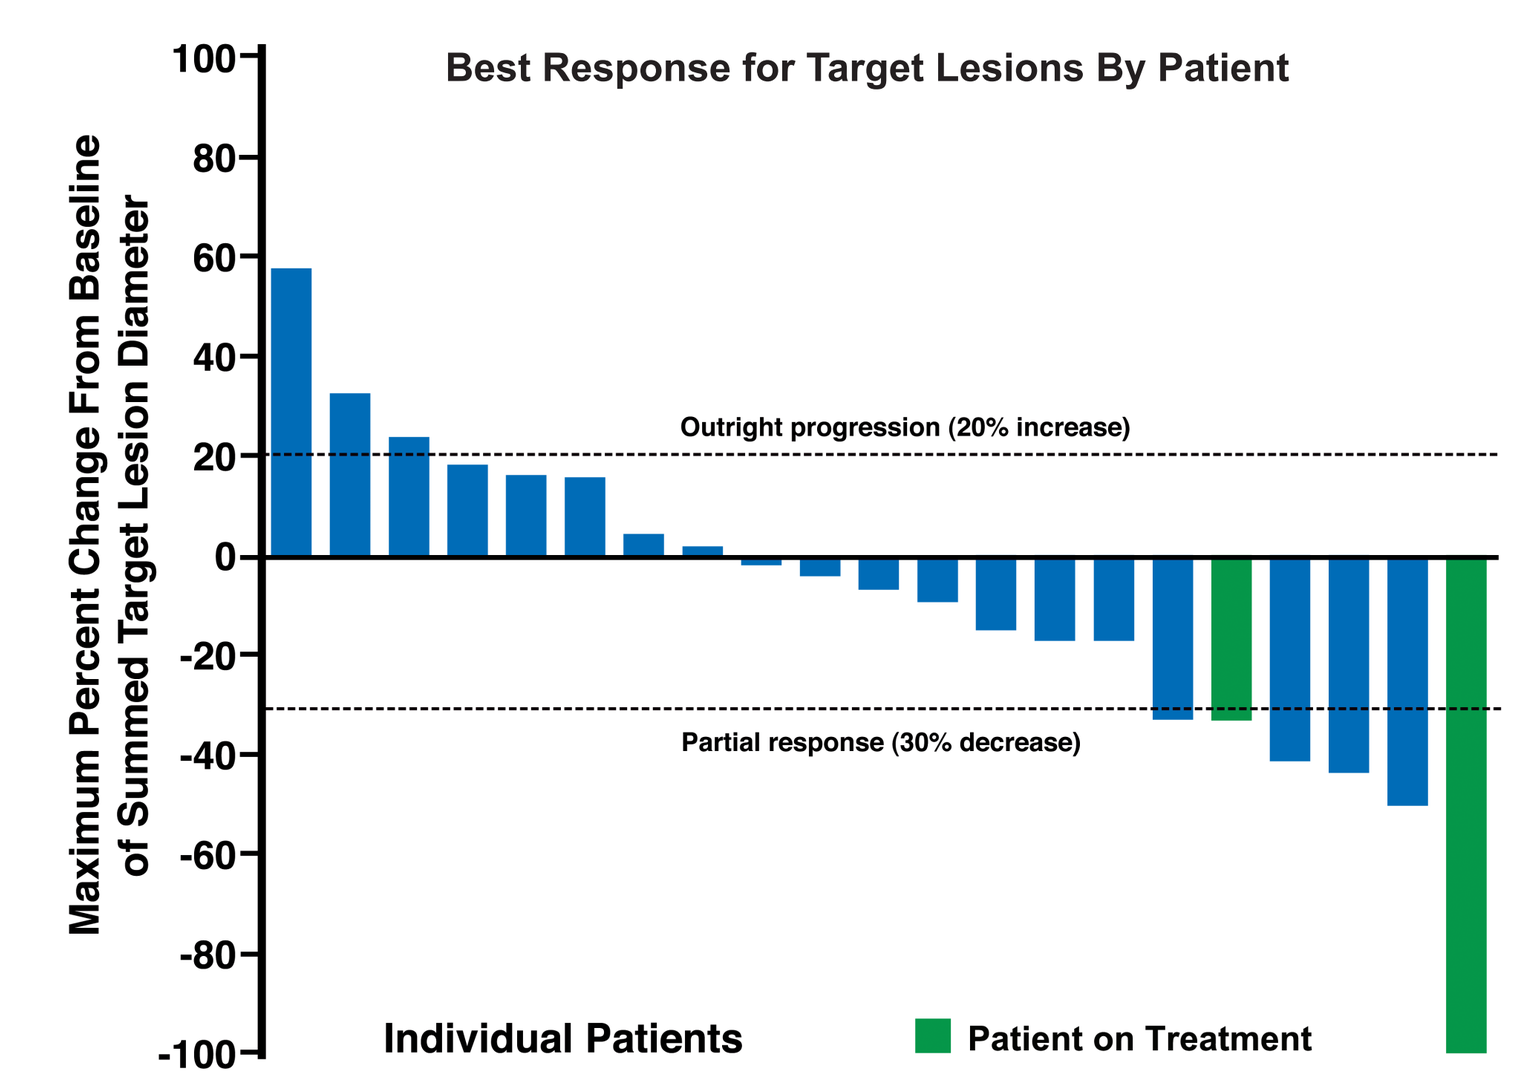
Best Response for Target Lesions By Patient
Maximum Percent Change From Baseline of Summed Target Lesion Diameter
100
80
60
40
20
0
-20
-40
-60
-80
-100
Outright progression (20% increase)
Partial response (30% decrease)
Individual Patients
Patient on Treatment

Survival

Patient Status: 2 continue on study treatment, 2 in follow-up, 19 deceased, 2 lost to follow-up.

Survival (days)	From Diagnosis	From Start of Treatment
Median	2495	166
Range	877-8746+	14-891+

CONCLUSION

This study demonstrates the feasibility of personalized cancer treatment for patients with progressing metastatic breast cancer using a first-of-its-kind highly multiplexed genomic and protein activation MP-rationalized treatment recommendation.

The multiplexed genomic-proteomic molecular profiling analysis and treatment recommendation were routinely delivered within 13-20 business days from biopsy, demonstrating feasibility of such an approach in a real-world clinical setting.

MP-based therapy selected by treatment selection committee was different in 100% (25/25) of cases compared to empiric choice selection.

Change in HER2 status (n=2) and HR status (n=2) significantly impacted treatment decision and likely response; supports the value of biopsy at the time of PD.

Patient-specific target driven treatment selection based on MP of a metastatic lesion provided clinical benefit for 13 of 25 (52%) heavily pretreated MBC pts. Thus, this approach merits further investigation in future studies.

References

Von Hoff DD, Stepheson JJ Jr., Rosen P.. et al. Pilot study using molecular profiling of patients' tumors to find potential targets and select treatments for their refractory cancers. JCO Nov. 20, 2010: 4877-4883.

Wulfkuhle JD, Edmiston KH, Liotta LA, Petricoin EF. Technology Insight: pharmacoproteomics for cancer-promises of patient-tailored medicine using protein microarrays. Nat Clin Pract Oncol. 2006 May;3(5):256-68.